Barriers to Best Management of Epilepsy

Marson AG[1], Bolan A[2], Mahon J[2], Little A[3], Dickson R[2], Boon P[4], Depondt C[1], Martikainen J[5], Ryvlin P[4], Kälviäinen R[4]

[1]International League Against Epilepsy (ILAE); [2]University of Liverpool; [3]European Federation of Neurological Associations (EFNA); [4]European Academy of Neurology (EAN); [5]University of Eastern Finland

VOT

EBC European Brain Council

Background

Epilepsy is a chronic neurological disorder characterized by repeated seizures (> 24 h apart); a single seizure with a high probability of seizure recurrence, or diagnosis of an epilepsy syndrome. The incidence of epilepsy in developed countries is approximately 50 per 100,000 individuals per year, with the greatest rates for infants and the elderly. In Europe epilepsy affects 6 million people (prevalence 8/1000) and the lifetime cumulative prevalence of epilepsy is 3 %. Despite the development of a new generation of antiepileptic drugs and surgical approaches to management, ~30% of patients are drug-refractory, with significant associated co-morbidities of depression, cognitive impairment and other neuropsychiatric diseases as well as increased risk of sudden unexpected death.

Methods

The epilepsy 'treatment gap', defined as the proportion of people with epilepsy who require treatment but do not receive it or receive inadequate treatment, has been proposed as a useful parameter to compare access to and quality of care for epilepsy patients across populations. The 'treatment gap' varies from 10-20 per cent in developed countries to 75 per cent in low-income countries. Stigma and discrimination related to epilepsy are also prevalent in Europe. In this study we assessed the epilepsy care pathway to identify the major barriers to achieving optimal treatment (both those needing research and better evidence to inform treatment decisions and those needing better organization of services).

Barriers to optimal treatment:

Investigation and management following an initial unprovoked seizure:

- **Scientific:** We cannot precisely predict who will have further seizures and we cannot prevent the development of epilepsy or the process of epileptogenesis.
- **Organizational:** Access to specialist services and assessment is variable even after a first tonic-clonic seizure. If the first symptoms are focal symptoms, the diagnosis may be delayed for years.

Treatment of patients who have a second (or more) unprovoked seizure:

- **Scientific:** For the great majority of people with epilepsy, we have no biomarkers that aid the choice of a specific drug for an individual. Indeed none may exist for currently available treatments, and drug choices are made based on knowledge (often inadequate) of effect in broad populations. As a result many patients undergo a period of trial and error in order to find the best treatment(s).
- **Organizational:** It is estimated that 70% of people could have their seizures fully controlled with timely and appropriate AED treatment. This would be achieved with early access to specialist assessment and comprehensive care including appropriate counselling and partnership with the patient. However, only 52% of people with epilepsy in the UK have their seizures fully controlled with AEDs. This indicates that currently available medical treatments are not used to their full potential.

Treatment of patients with epilepsy that is refractory to medical treatments:

- **Scientific:** We have no biomarkers to allow early identification of patients destined to be refractory and who would benefit from epilepsy surgery or neuromodulation.
- **Organizational:** There is substantial underutilization of the treatment options for refractory epilepsy. The mean delay between first seizures and epilepsy surgery is still nearly 20 years for the majority and it is estimated that only 40% of refractory patients get comprehensive diagnostic evaluation. As soon as patients are diagnosed as having refractory epilepsy, they should all be referred for comprehensive diagnostic evaluation to a tertiary epilepsy centre

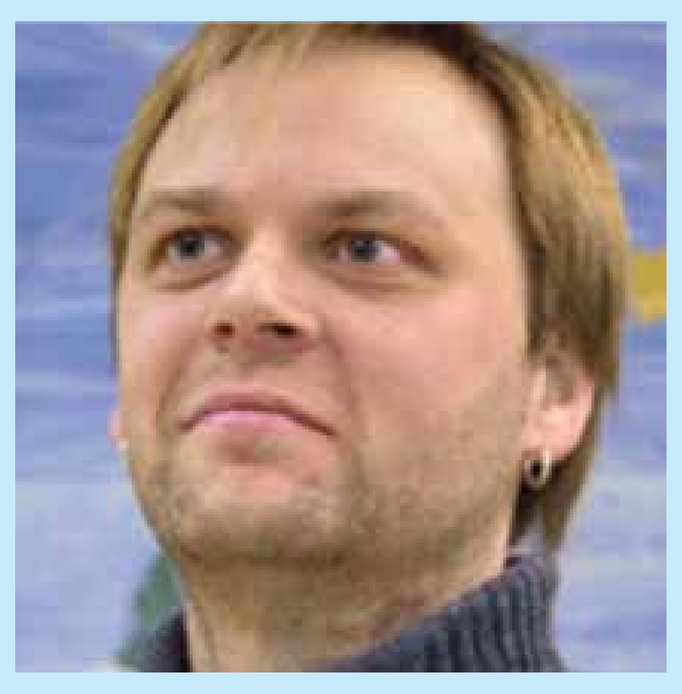

Juha Karjula, a 38 year-old kindergarten teacher, obtained his university Master's degree despite having refractory epilepsy. He was diagnosed with focal epilepsy as a high school student, after having his first tonic-clonic seizure. Despite of detailed investigations, the cause of his epilepsy has remained unknown. For years his seizures were not adequately controlled with different AEDs, which were used either as monotherapy or in combination therapy. Some AEDs also resulted in unbearable side-effects, such as speech problems. However, with comprehensive evaluation and with arduous successive treatment attempts, seizure control was achieved in 2011. At the moment Juha is using a combination of two AEDs. He is seizure-free and has no significant side-effects of his current treatment.

In his experience, it is important to have good communication between a person with epilepsy and their neurologist. If needed, a support person or a family member, someone who knows a patient's everyday life and wishes, could act as 'interpreter' between the doctor and the patient to ensure the patient is understood. In this way Juha had good interaction with his doctor and found a way for mutual understanding with his doctor about how he wants to be treated.

Text: Sari Tervonen, Photo: Hannu Miettinen IBE/Finland

Recommendations

- Raise public awareness (campaign) and awareness among health care professional (GPs, general physicians, emergency medicine)
- Provide specialist epilepsy training (neurologists, specialist nurses)
- Link emergency and GP services with epilepsy / seizure services to ensure rapid access to expertise following first seizures(s).
- Provide access to specialist expertise for refractory patients
- Provide timely access to epilepsy surgery services
- Provide peer-support in collaboration with patient organizations
- Invest in research

Conclusions

Closing the epilepsy treatment gap will require a multifaceted approach, including raising awareness of epilepsy and the effectiveness of treatments in the general population and among those working in health services; training healthcare professionals with skills to diagnose and manage epilepsy. Service need to be well-coordinated, networked and accessible allowing management from first seizure through to complex epilepsy surgery, whilst also taking into account and managing co-morbidities.

References:

International Bureau for Epilepsy. EPILEPSY IN THE WHO EUROPEAN REGION: Fostering Epilepsy Care in Europe. EPILEPSY out of the Shadows. Available at: http://www.ibeepilepsy.org/downloads/EURO%20Report%20160510.pdf. ; Jozwiak S, et al. 2017. WONOEP appraisal: Development of epilepsy biomarkers-What we can learn from our patients? Epilepsia.; Engel J Jr. 2016. What can we do for people with drug-resistant epilepsy? The 2016 Wartenberg Lecture. Neurology.

Acknowledgements:

This work was supported by and UCB Biopharma SPRL. We would like to thank Peter Dedeken for the contribution to this work.
A digital version of the poster and other supporting documents are available here:
http://www.braincouncil.eu/activities/projects/the-value-of-treatment/epilepsy

ean the home of neurology

UNIVERSITY OF LIVERPOOL

EFNA European Federation of Neurological Associations

Epilepsy Alliance Europe under the auspices of: ILAE IBE

Cost savings and improved patient outcomes from best management of epilepsy

Marson AG[1], Bolan A[2], Mahon J[2], Little A[3], Dickson R[2], Boon P[4], Depondt C[1], Martikainen J[5], Ryvlin P[4], Kälviäinen R[4]

[1]International League Against Epilepsy (ILAE); [2]University of Liverpool; [3]European Federation of Neurological Associations (EFNA); [4]European Academy of Neurology (EAN); [5]University of Eastern Finland

Background

Clinical guidelines have been published in the UK recommending that patients with epilepsy should be treated by specialists and rapidly offered alternative treatments to anti-epileptic drugs (AEDs) if they are not achieving remission by 2 years.[1] The aim of this research was to assess how health outcomes for patients in the UK NHS, and the resource use associated with seizures and complications, would change if care were delivered in line with the NICE Guideline (`best management') instead of how patients with epilepsy are currently being managed (`current management'). Best management was simplified into the following key principles of care:

- Patients are referred to an epilepsy specialist after two seizures
- Specialists select an initial AED and monitor and adapt treatment as required
- Patients not in remission at 2 years are assessed for surgery and those eligible are offered surgery
- Patients ineligible for surgery are assessed for vagal nerve stimulation (VNS) and those eligible are offered VNS.

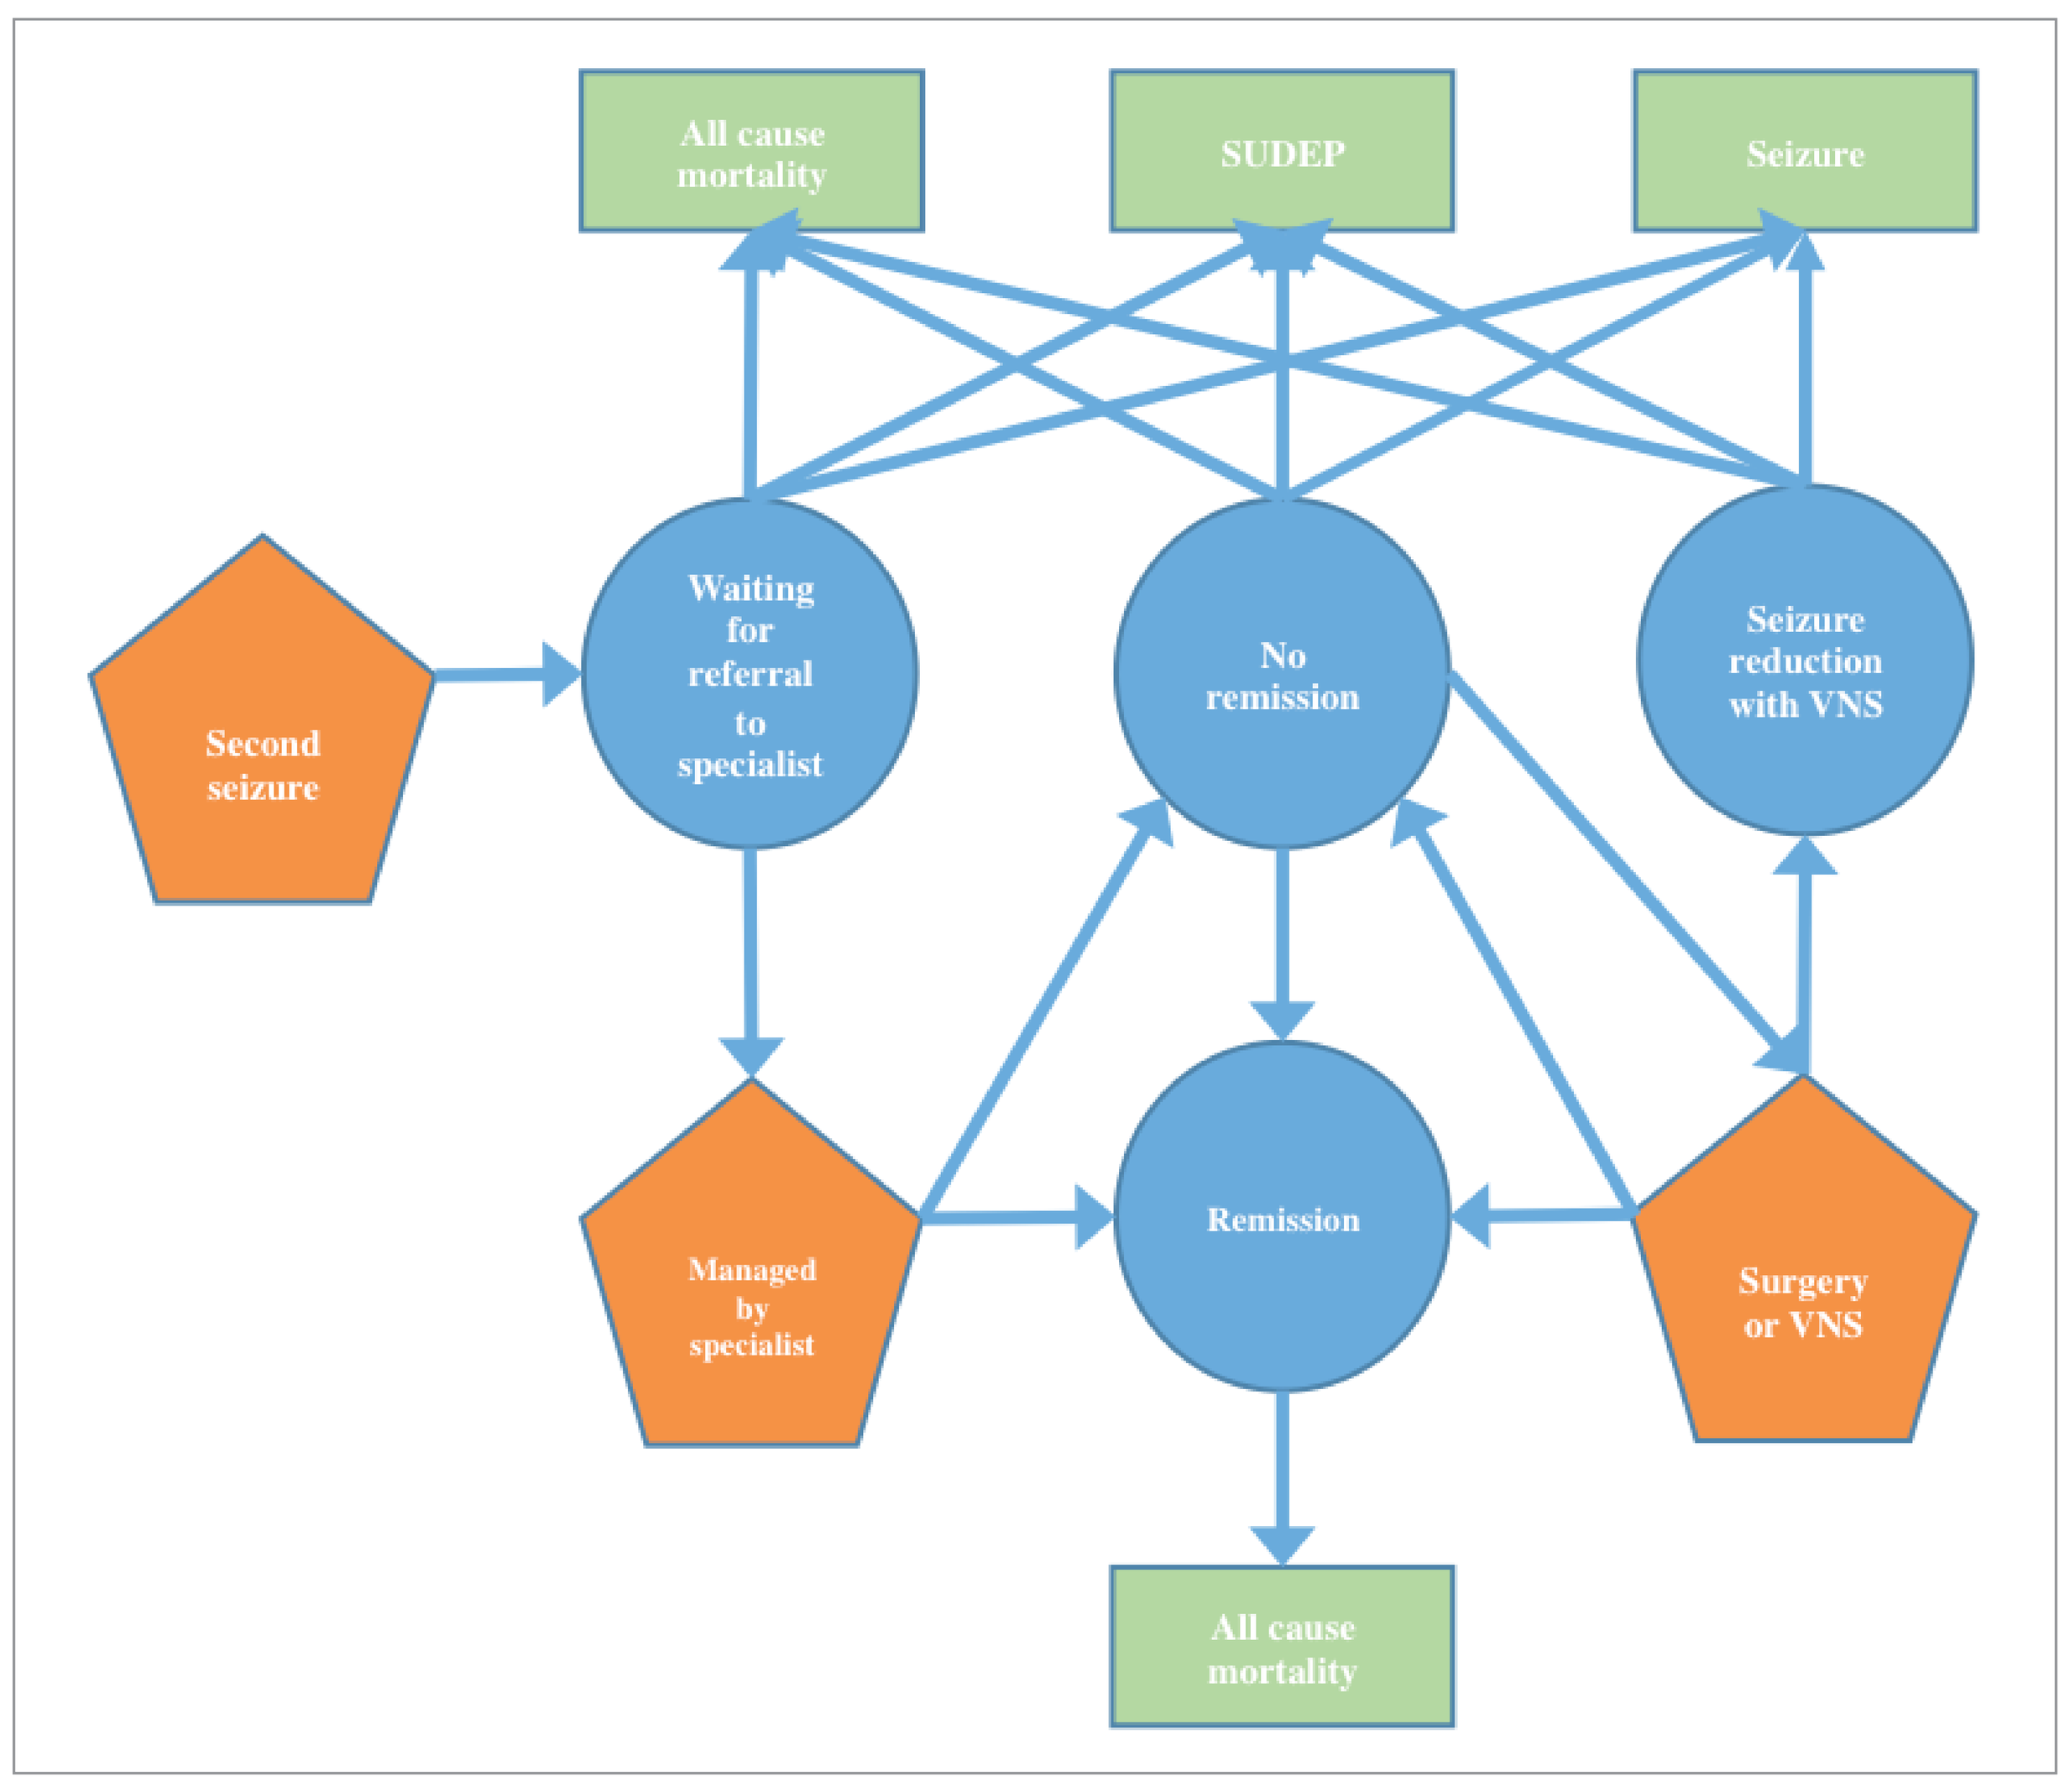
All cause mortality
SUDEP
Seizure
Second seizure
Waiting for referral to specialist
No remission
Seizure reduction with VNS
Managed by specialist
Remission
Surgery or VNS
All cause mortality

Figure 1: Simplification of model pathway

Methods

A Markov model with a 25 year time horizon and one week cycle was constructed to explore the impact on the patient and healthcare provider (the NHS) from moving from current management of people with epilepsy to best management. The model pathway starts from second seizure (patient age 30), moves to management by a specialist (treatment with AEDs) and finally goes to surgery or provision of VNS. In each cycle there is a probability a patient has a seizure (non-convulsive, convulsive status epilepticus or refractory convulsive status epilepticus), or dies from SUDEP (sudden unexpected death in epilepsy) or from any cause (Figure 1).

Estimates of remission and seizure rates under best and current management with AEDs were derived from the SANAD (Standard And New Antiepileptic Drugs) trial data.2 SANAD baseline data, other published sources and clinical assumption were used to determine remaining transition probabilities. Costs were incurred from the perspective of the NHS. Only costs associated with seizures and their complications were considered. Utility values were identified from the literature for neurological damage and for people with epilepsy that were not in remission.

In all cases, the model was constructed conservatively to provide a lower estimate of the benefits of best management. For example the costs of neurological damage from seizures were not considered in the base case.

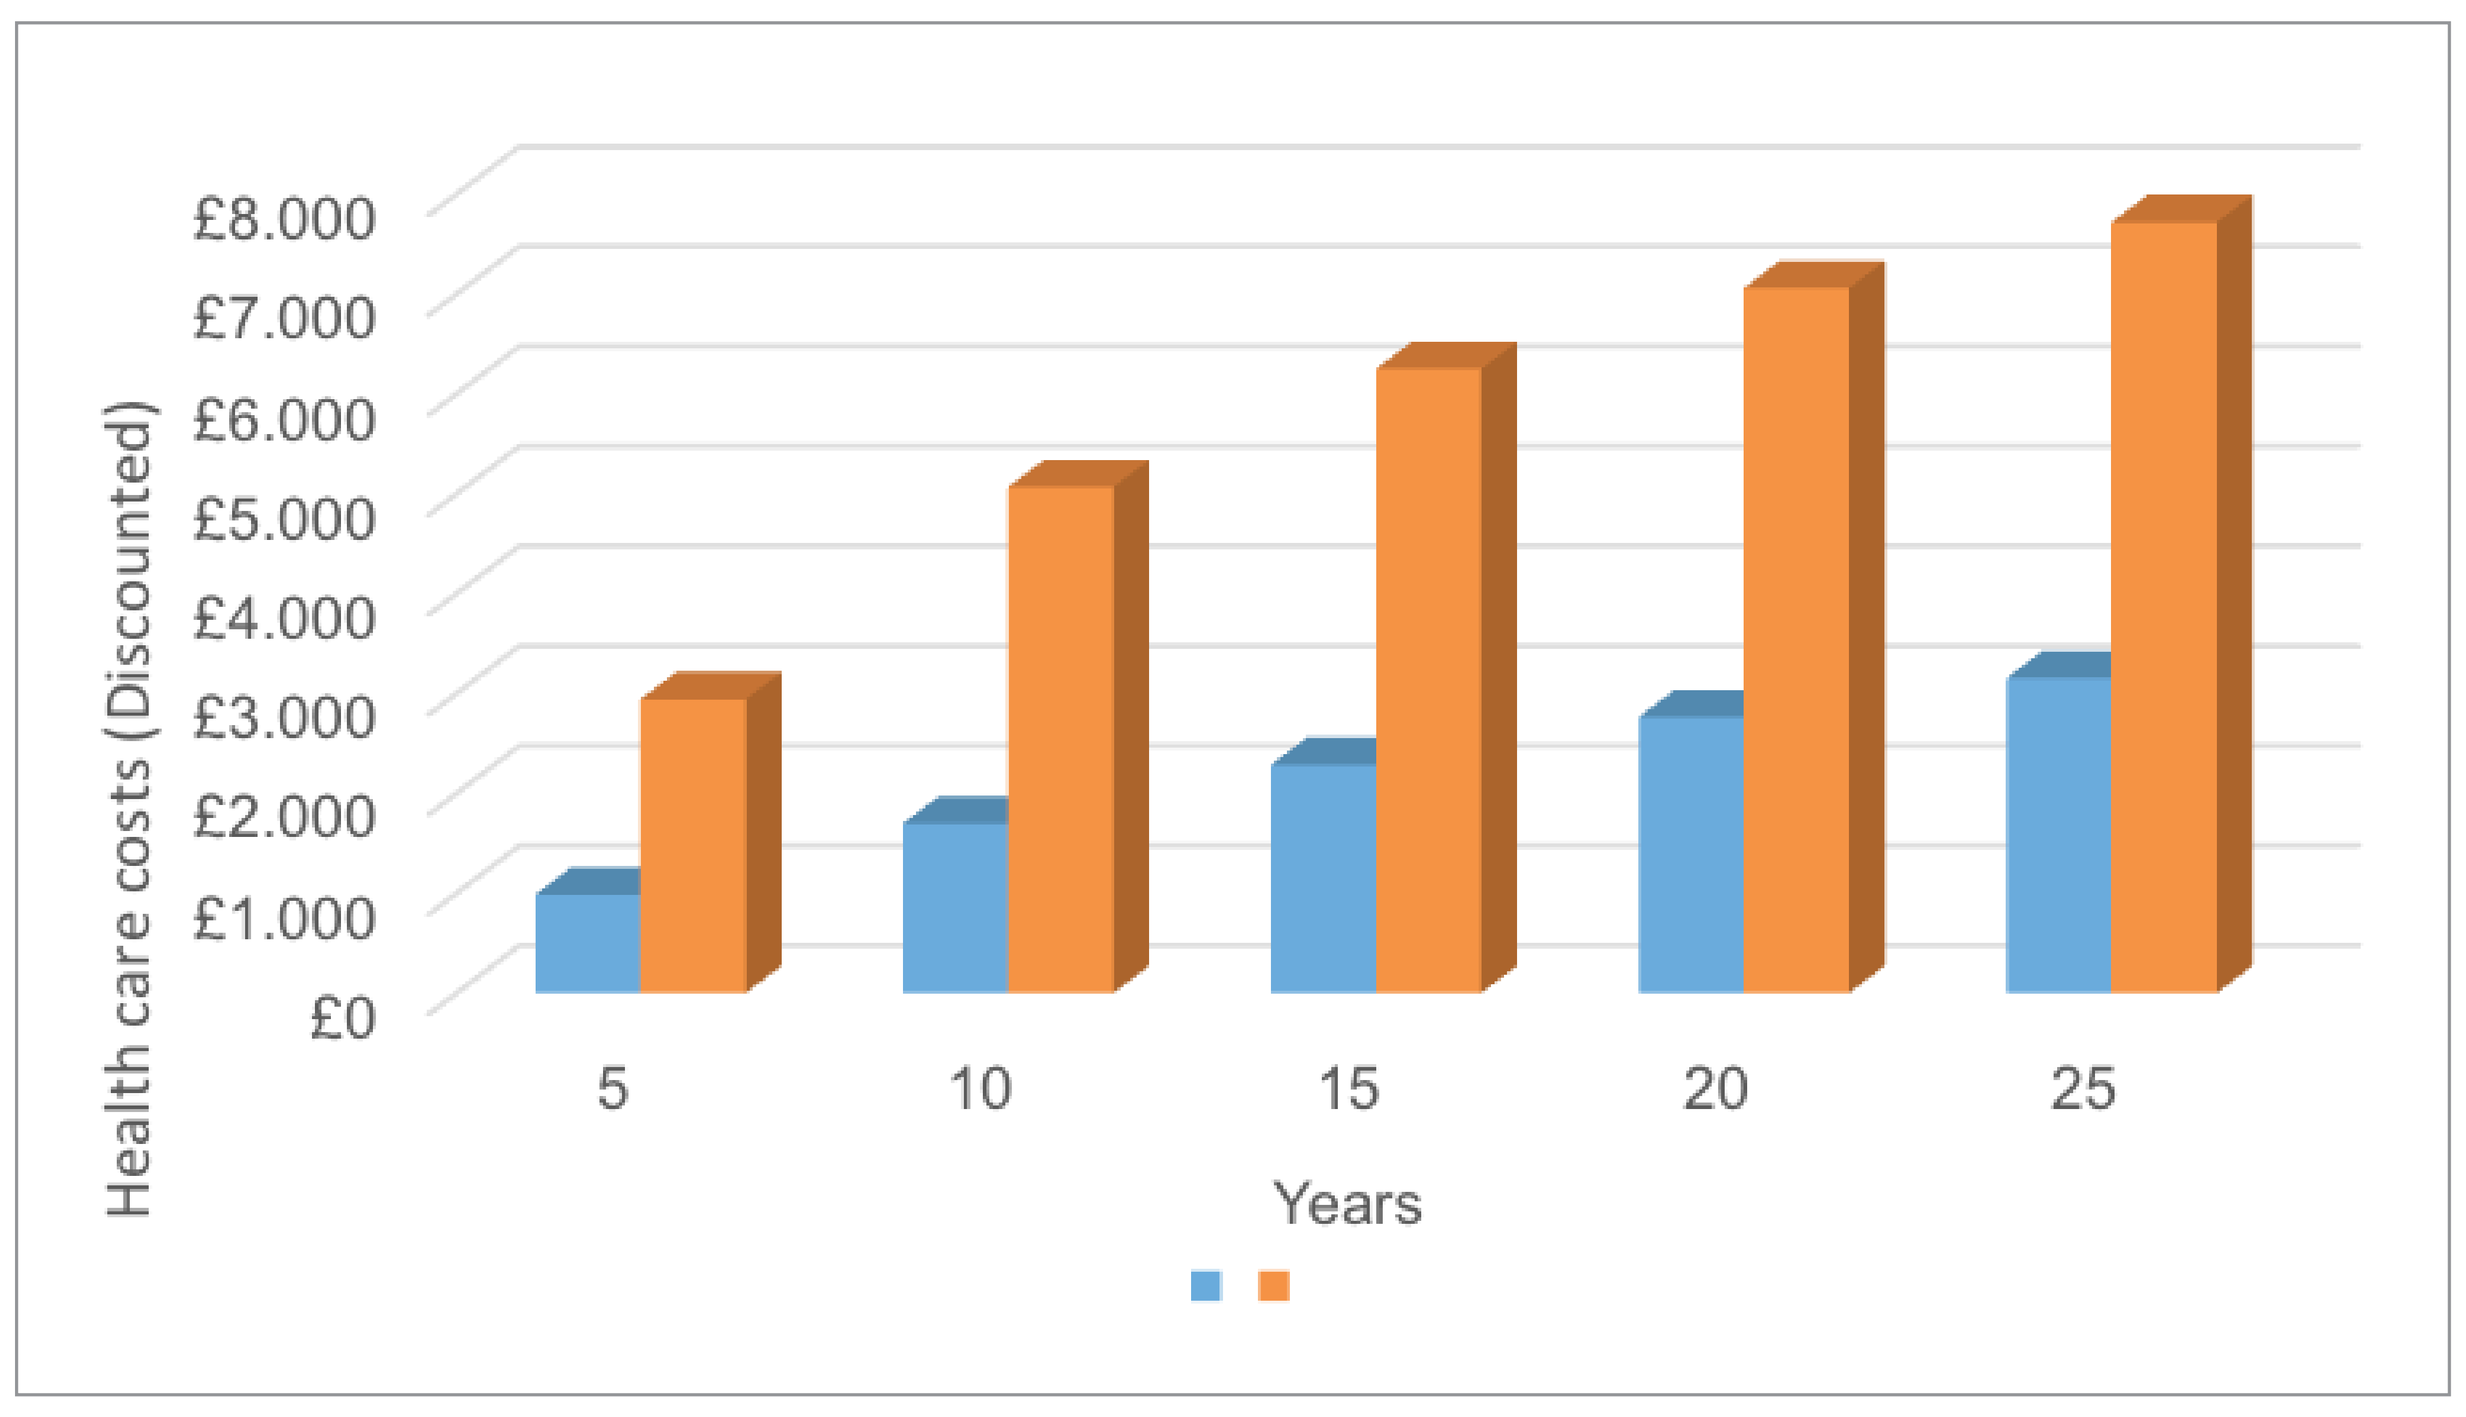
£8.000
£7.000
£6.000
£5.000
£4.000
£3.000
£2.000
£1.000
£0
Health care costs (Discounted)
5
10
15
20
25
Years

Figure 2: Healthcare costs per person over 25 years with best and current management

Results

Initial results from the model suggest that significant improvements in patient outcomes and an associated reduction in costs could be achieved with a move from current management to best management in the UK NHS. The model suggests that, over 25 years, best management would generate cost savings of £4,565 (discounted) or £6,033 (undiscounted) per patient from treating seizures and associated complications. There would also be a quality adjusted life year (QALY) gain per patient of 1.05 (discounted) or 1.62 (undiscounted). The cost and QALY savings over 5 year periods up to 25 years are shown in Figures 1 and 2 respectively.

The costs savings are generated from a reduction in convulsive seizures, which with best management are reduced from an average of 8.2 per patient to 3.5 per patient over 25 years. This in turn results in a 56.7% reduction in the costs (undiscounted) of treating convulsive seizures, such that costs fall from £10,605 to £4,589 per person over 25 years. With a 67.7% reduction in patients with neurological damage (from 25.5 to 8.3 per 1,000 patients), there would also be a significant reduction in the costs of treatment and potential ongoing care for this group; these additional costs have not been included in the modelling. The QALY gains are generated from a 48.7% reduction in epilepsy related mortality (SUDEP or death from seizure) over 25 years (mortality falls from 75.2 to 38.5 per 1,000 patients), the reduction in patients with long-term neurological damage and from an increase in patients being seizure free and becoming seizure free earlier. By 10 years the model predicts that, with best management, 61.0% of patients will be alive and in remission. With current management only 33.0% will be alive and seizure free. With best management at 10 years, a further 11.6% will have had seizure reduction with VNS as opposed to only 0.2% with current management.

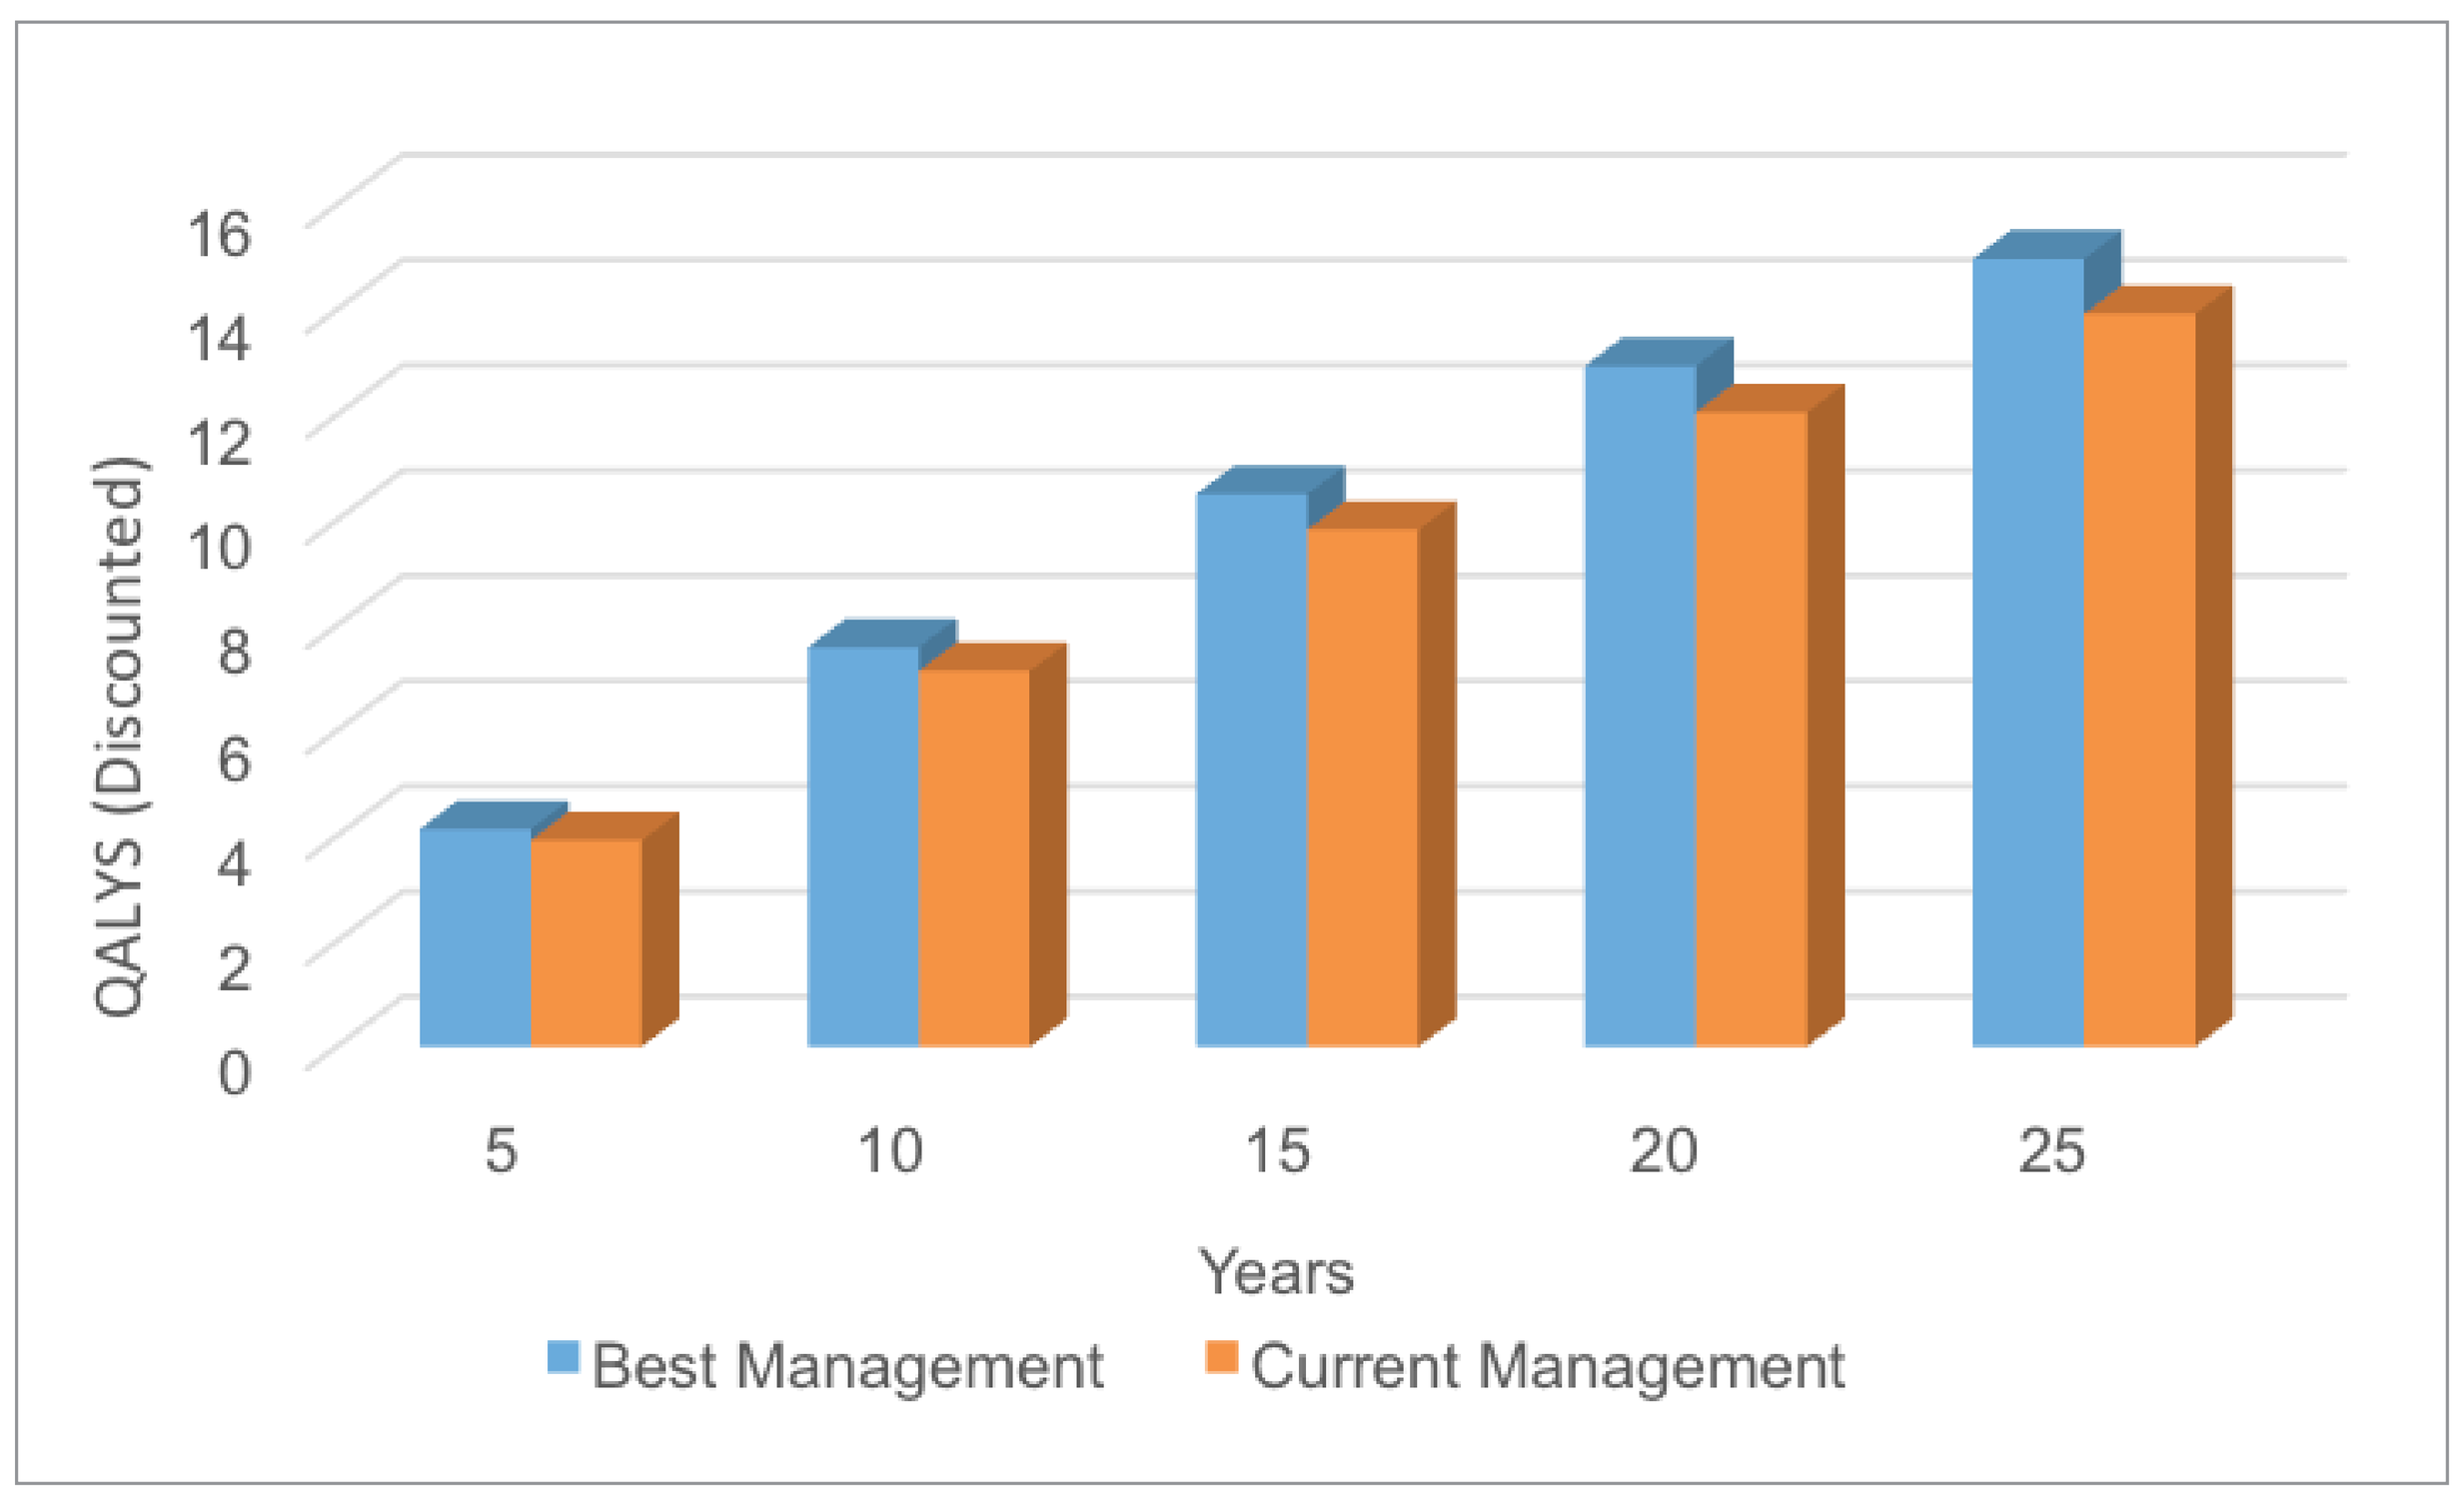
16
14
12
10
8
6
4
2
0
QALYS (Discounted)
5
10
15
20
25
Years
Best Management
Current Management

Figure 3: QALYs per person over 25 years with best and current management

Conclusions

Under a conservative set of assumptions and utilising the seizure experience of patients prior to joining the SANAD trial and seizure reduction seen whilst on the trial, the results of our analysis suggest that the improvement in patient outcomes and reduction in healthcare costs from current to best management of epilepsy would be significant. Achieving best management would not be without cost. However, if NICE values a QALY at £20,000, best management would be considered to offer good value for money to the UK NHS (due to the expected costs savings and QALY gains) provided that it costs less than an additional £25,500 per patient to achieve.

References:

National Institute for Health and Care Excellence. 2016. Epilepsies: diagnosis and management (CG137). https://www.nice.org.uk/guidance/cg137; Marson AG et al. 2007. The SANAD study of effectiveness of valproate, lamotrigine, or topiramate for generalised and unclassifiable epilepsy: an unblinded randomised controlled trial. Lancet.

Acknowledgements:

This work was supported by UCB Biopharma SPRL. We would like to thank Peter Dedeken (UCB Biopharma SPRL) for the contribution to this work. A digital version of the poster and other supporting documents are available here:
http://www.braincouncil.eu/activities/projects/the-value-of-treatment/epilepsy